Phase IIb Randomized Study of CPX-351 vs. Conventional Cytarabine + Daunorubicin in Newly Diagnosed AML Patients Aged 60-75: Safety Report

J. Lancet[1], E. Feldman[2], J. Cortes[3], D. Hogge[4], T. Kovacsovics[5], M. Tallman[6], L. Damon[7], S. Solomon[8], J. Kolitz[9], R. Komrokji[1], M. Chiarella[10], A. Louie[10]

[1]H. Lee Moffitt Cancer Center, Tampa, FL, [2]Weill Cornell Medical Center, New York, NY, [3]MD Anderson Cancer Center, Houston, TX, [4]British Columbia Cancer Agency, Vancouver, BC, [5]Oregon Health and Science University, Portland, OR, [6]Northwestern University-Feinberg School of Medicine, Chicago, IL, [7]University of California Medical Center, San Francisco, CA, [8]Blood and Marrow Transplant Group of Georgia, Atlanta, GA, [9]Monter Cancer Center, Lake Success, NY, [10]Celator Pharmaceuticals Inc., Princeton, NJ

Introduction

CPX-351 is a liposomal formulation of cytarabine and daunorubicin encapsulated at a 5:1 molar ratio, that has been shown to be synergistic/non-antagonistic across multiple solid tumor and leukemic cell lines. A phase I dose escalating study conducted in patients with refractory/relapsed acute leukemia defined the MTD (101 units/m^2, Day 1, 3 and 5), confirmed that the 5:1 molar ratio could be maintained in the plasma for over 48 hours and that both drugs and their active metabolites had prolonged half-lives and full bioavailability. The most frequent and clinically important adverse events were those related to neutropenia and thrombocytopenia and included febrile neutropenia, bacteremia, sepsis and pneumonia. Aplasia and complete remissions were observed in patients following multiple relapses, first relapse after short duration first CR and in one case after primary induction failure. Remissions were observed at dose levels less than 50% of the MTD, suggesting potent anti-leukemic activity. A randomized phase II study was initiated to compare the safety and efficacy of CPX-351 directly against conventional cytarabine and daunorubicin (“7+3” regimen).

Study Schema

- Newly diagnosed AML
- Age 60 - 75 years
- High risk and standard risk leukemia based on karotype, AHD and age
- PS 0-2
- LVEF>50%
- Up to 2 inductions and 2 consolidations

CPX-351 100 units/m^2 (80 pt)

86pts→57pts

“7+3” Regimen (40 pt)

41pts→28pts

Methods

Patients with known favorable cytogenetics, active secondary malignancies, active bacterial infections and those with prior overall cumulative exposure >368mg/m^2 of daunorubicin were excluded. Primary induction failures on the control arm could be crossed-over to receive CPX-351 or be discontinued from the study. Patients were automatically assigned to the high risk group if they had secondary AML or were ≥70 years of age, or had ≥3 chromosomal abnormalities. All other patients were assigned to the standard risk group. One hundred twenty seven patients were randomized: 86 to CPX-351 treatment and 41 to “7+3” control. This interim analysis presents data collected by 15 August 2009 for the first 85 patients enrolled.

Demographics

The average patient was male (70.6%), white (90.6%), 68 years of age, had ECOG performance status of 0-1 (83.5%) and had de novo AML (66%). The majority of patients were assigned to the high risk group (57.6%). Stratification successfully assigned high risk and standard risk patients equally to the CPX-351 arm and the control arm of the study. The CPX-351 arm had more patients with ECOG PS=2 (21.1% vs 7.1%), <3 chromosomal abnormalities (43.9% vs 28.6%) and had a higher proportion of patients with baseline WBC>20,000/μL (22.9% vs 14.3%).

Conflict of Interest Disclosure

M. Chiarella and A. Louie are employees of Celator Pharmaceuticals, Inc.

Table 1: Demographics

	Regimen: / Patients:	CPX-351 n=57	"7+3" n=28	n=85
Gender	Male	42 (73.7%)	18 (64.3%)	60 (70.6%)
	Female	15 (26.3%)	10 (35.7%)	25 (29.4%)
Age (yr)	Mean	67.6 (4.91)	68.0 (4.75)	67.8 (4.83)
	Median	68	68	68
	60-65 yrs	19 (33.3%)	9 (32.1%)	28 (32.9%)
	66-70 yrs	18 (31.6%)	9 (32.1%)	27 (31.8%)
	>70 yrs	20 (35.1%)	10 (35.7%)	30 (35.3%)
Race	White	50 (87.7%)	27 (96.4%)	77 (90.6%)
	Black/African American	5 (8.8%)	1 (3.6%)	6 (7.1%)
	Other	2 (3.5%)	0 (0.0%)	2 (2.4%)
ECOG	0	18 (31.6%)	11 (39.3%)	29 (34.1%)
	1	27 (47.4%)	15 (53.6%)	42 (49.4%)
	2	**12 (21.1%)**	2 (7.1%)	14 (16.5%)
AML type	De novo AML	38 (66.7%)	18 (64.3%)	56 (65.9%)
	Secondary AML	19 (33.3%)	10 (35.7%)	29 (34.1%)
Cytogenetics	≥3	4 (7.0%)	3 (10.7%)	7 (8.2%)
	<3	**25 (43.9%)**	8 (28.6%)	33 (38.8%)
	unknown	28 (49.1%)	17 (60.7%)	45 (52.9%)
Baseline WBC	<20K/μL	44 (77.2%)	24 (85.7%)	68 (80.0%)
	20-100K/μL	**12 (21.1%)**	4 (14.3%)	16 (18.8%)
	>100K	**1 (1.8%)**	0 (0.0%)	1 (1.2%)
Risk Group	Standard Risk	24 (42.1%)	12 (42.9%)	36 (42.4%)
	High Risk	33 (57.9%)	16 (57.1%)	49 (57.6%)

Efficacy

- Clearance of leukemia (CR and CRi) occurred in 61.4% of CPX-351 treated patients and in 50% of “7+3” control arm patients (p=ns).

Table 2: Efficacy

Regimen: / Patients:	CPX-351 n=57	"7+3" n=28
	OVERALL	
CR (morphologic)	23 (40.4%)	11 (39.3%)
CRi	12 (21.1%)	3 (10.7%)
CR+CRi	35 (61.4%)	14 (50.0%)
	STRATIFICATION GROUP	
Overall Risk		
Standard Risk	16/24 (66.7%)	6/12 (50.0%)
High Risk	19/33 (57.6%)	8/16 (50.0%)
Chromosomal Abnormalities Randomization		
≥3	3/4 (75.0%)	1/3 (33.3%)
<3	14/25 (56.0%)	5/8 (62.5%)
unknown	18/28 (64.3%)	8/17 (47.1%)
Age Group		
60-65 yrs	12/19 (63.2%)	4/9 (44.4%)
66-70yrs	11/18 (61.1%)	5/9 (55.6%)
>70yrs	12/20 (60.0%)	5/10 (50.0%)
AML Type		
De novo AML	27/38 (71.1%)	11/18 (61.1%)
Secondary AML	8/19 (42.1%)	3/10 (30.0%)

- When examined by overall risk group, number of chromosomal abnormalities, age group and AML type, CPX-351 appears to have equal or greater complete remission rate when compared to the control arm.
- For CPX-351: 30 of the 35 (86%) complete responders achieved remission after one induction.
- For “7+3”: 10 of the 14 (71%) complete responders achieved remission after one induction.

Safety

Table 4: Mortality

Regimen: / Patients:	CPX-351 n=57	"7+3" n=28
Mortality by Period:		
≤7 days	0 (0.0%)	0 (0.0%)
8-30 days	3 (5.3%)	1 (3.6%)
31-60 days	0 (0.0%)	0 (0.0%)
61-90 days	2 (3.5%)	1 (3.6%)

- Deaths within the first 30 days were similar in both arms of the study (5.3% vs 3.6%).

Serious Adverse Events (45.6% vs 21.4%) appeared to be more frequent in the CPX-351 arm and were dominated by infection-related AEs. Of 34 patients in the CPX-351 arm, 14 had only infection related SAEs, 14 had non-infection related SAEs and 6 patients had both infection and non-infection related SAEs. Similarly of 13 patients in the control arm, 4 had infection only SAEs, 7 had non-infection SAEs, and 2 had both types of events.

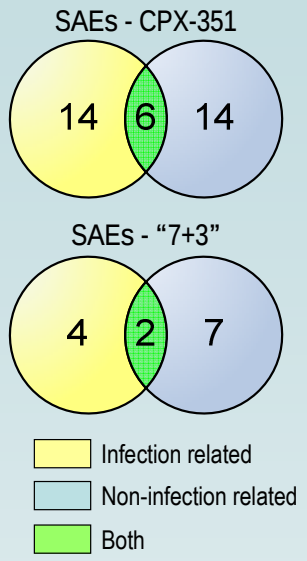

SAEs - CPX-351
14
6
14
SAEs - “7+3”
4
2
7
Infection related
Non-infection related
Both

- All 85 patients experienced at least one adverse event.
- The overall frequency of adverse events and Grade 3 and 4 adverse events for the gastro-intestinal, metabolic, skin, respiratory, renal, vascular and nervous systems were comparable.
- With small numbers of events reported, CPX-351 treatment appeared to be associated with more bacterial pneumonias Grade 3 and 4 (15.8% vs 7.1%), urinary tract infections Grade 3 and 4 (5.3% vs 0%) and mental status changes Grade 3 and 4 (5.3% vs 0%).
- Five patients treated with CPX-351 experienced left ventricular dysfunction with clinically apparent CHF reported in 3 patients (2 Grade 3 and 1 Grade 4). The “7+3” group had a single patient with left ventricular dysfunction (Grade 1/2).

Aplasia Recovery

CPX-351 required longer time to achieve peripheral blood count recovery. Median time for recovery of ANC to >500/μL was 37.5 days for CPX-351 and 28 days for “7+3”. Median time for recovery of platelets to >20,000/μL was 28 days for CPX-351 and 21 days for “7+3”.

Crossover

Seven of the 28 patients randomized to control treatment were crossed over to receive CPX-351; 3 with de novo AML and 4 with secondary AML.CPX-351 treatment produced clearance of leukemia in 5 patients resulting in 3 CRs: 1 with de novo AML and 2 with secondary AML. These findings are encouraging and suggest that CPX-351 may be active in patients with primary induction failure.

Table 3: Adverse Events

Regimen:	CPX-351 n=57								"7+3" n=28							
Adverse Events:	Grades 1 and 2		Grade 3		Grade 4		Incidence Grades 3 and 4		Grades 1 and 2		Grade 3		Grade 4		Incidence Grades 3 and 4	
	n	%	n	%	n	%	n	%	n	%	n	%	n	%	n	%
Cardiac																
Congestive Heart Failure	2	(3.5%)	2	(3.5%)	1	(1.8%)	3	(5.3%)	1	(3.6%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Acute MI	0	(0.0%)	0	(0.0%)	1	(1.8%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	1	(3.6%)	1	(3.6%)
Gastrointestinal																
Nausea	31	(54.4%)	1	(1.8%)	0	(0.0%)	1	(1.8%)	16	(57.1%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Vomitting	10	(17.5%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	8	(28.6%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Stomatitis	12	(21.1%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	6	(21.4%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Diarrhea	29	(50.9%)	6	(10.5%)	0	(0.0%)	6	(10.5%)	16	(57.1%)	2	(7.1%)	0	(0.0%)	2	(7.1%)
Gastro-Intestinal Necrosis	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	1	(3.6%)	1	(3.6%)
Intestinal Perforation	0	(0.0%)	0	(0.0%)	1	(1.8%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Small Intestinal Obstruction	0	(0.0%)	2	(3.5%)	0	(0.0%)	2	(3.5%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Infections																
Bacteremia	3	(5.3%)	12	(21.1%)	0	(0.0%)	12	(21.1%)	2	(7.1%)	4	(14.3%)	0	(0.0%)	4	(14.3%)
Pneumonia	5	(8.8%)	9	(15.8%)	0	(0.0%)	9	(15.8%)	1	(3.6%)	2	(7.1%)	0	(0.0%)	2	(7.1%)
Sepsis	1	(1.8%)	1	(1.8%)	2	(3.5%)	3	(5.3%)	0	(0.0%)	1	(3.6%)	1	(3.6%)	2	(7.1%)
Pneumonia-Fungal	0	(0.0%)	4	(7.0%)	0	(0.0%)	4	(7.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Urinary Tract Infection	0	(0.0%)	3	(5.3%)	0	(0.0%)	3	(5.3%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Fungal Retinitis	0	(0.0%)	0	(0.0%)	1	(1.8%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Metabolism																
Anorexia/Reduction in Apetite	19	(33.3%)	2	(3.5%)	0	(0.0%)	2	(3.5%)	9	(32.1%)	2	(7.1%)	0	(0.0%)	2	(7.1%)
Skin																
Rash	30	(52.6%)	3	(5.3%)	0	(0.0%)	3	(5.3%)	16	(57.1%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Renal																
Acute Renal Failure	4	(7.0%)	2	(3.5%)	2	(3.5%)	4	(7.0%)	0	(0.0%)	2	(7.1%)	0	(0.0%)	2	(7.1%)
Respiratory																
Dyspnea	11	(19.3%)	1	(1.8%)	2	(3.5%)	3	(5.3%)	3	(10.7%)	1	(3.6%)	0	(0.0%)	1	(3.6%)
ARDS	0	(0.0%)	0	(0.0%)	1	(1.8%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Respiratory Failure	2	(3.5%)	1	(1.8%)	1	(1.8%)	2	(3.5%)	2	(7.1%)	1	(3.6%)	0	(0.0%)	1	(3.6%)
Vascular																
Hypertension	8	(14.0%)	1	(1.8%)	0	(0.0%)	1	(1.8%)	3	(10.7%)	2	(7.1%)	0	(0.0%)	2	(7.1%)
Hypotension	8	(14.0%)	1	(1.8%)	1	(1.8%)	2	(3.5%)	6	(21.4%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Psychiatric																
Psychotic Disorder	0	(0.0%)	1	(1.8%)	0	(0.0%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	1	(3.6%)	1	(3.6%)
Hallucination	7	(12.3%)	1	(1.8%)	0	(0.0%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Mental Status Changes	1	(1.8%)	3	(5.3%)	0	(0.0%)	3	(5.3%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)
Nervous System																
Subarachnoid Hemorrhage	0	(0.0%)	1	(1.8%)	0	(0.0%)	1	(1.8%)	0	(0.0%)	0	(0.0%)	0	(0.0%)	0	(0.0%)

Discussion and Conclusions

CPX-351 encapsulates, maintains and delivers to leukemic cells cytarabine and daunorubicin at a 5:1 synergistic molar ratio. Clinical studies to date have demonstrated potent activity against relapsed AML, with an acceptable safety profile. This randomized phase II study evaluated CPX-351 versus “7+3” in newly diagnosed elderly patients with AML and achieved clearance of leukemia (CR or CRi) in 61.4% of treated patients, an encouraging sign of anti-leukemic activity consistent with earlier clinical findings. There was evidence of prolonged cytopenias, some additional Grade 3 infectious adverse events and a few cases of cardiac toxicity in an otherwise well tolerated treatment. Final results from this study, including CR duration and survival at one year, will be available by late 2010, along with findings from an ongoing randomized study in patients with AML in first relapse. An interim analysis of data from the first randomized phase II study of CPX-351 in newly diagnosed elderly patients with AML demonstrated encouraging anti-leukemic activity with 61.4% of patients achieving CR or CRi.

Presented at American Society of Hematology Annual Meeting, 5-8 December 2009, New Orleans, LA, Abstract# 1033